Evaluation of Global Improvement and Lifetime Costs of Treating Tardive Dyskinesia Shows Treatment with Valbenazine Dominates Deutetrabenazine

Michael Ganz,[1] Sean Stern,[2] Ameya Chavan,[3] Michael Serbin,[4] Chuck Yonan[4]

[1]Evidera, Waltham, MA; [2]Evidera, San Francisco, CA; [3]Evidera, Bethesda, MD; [4]Neurocrine Biosciences, Inc., San Diego, CA

INTRODUCTION

- Tardive dyskinesia (TD) is an involuntary movement disorder associated with prolonged exposure to dopamine receptor-blocking agents, such as antipsychotics[1-3]
- TD can lead to physical and psychological impairment, social isolation, work-related disability, and decreased quality of life[4,5]
- Valbenazine (VBZ; INGREZZA®) and deutetrabenazine (DTBZ; AUSTEDO®) are vesicular monoamine transporter 2 (VMAT2) inhibitors approved for treatment of TD in adults
- The objective of the current study was to evaluate clinical and economic outcomes associated with VBZ and DTBZ compared with no treatment in patients with TD, using a model that
 - Assessed global improvements after 1 year of treatment, as judged separately by clinicians and patients
 - Measured the impact of TD on management of the underlying psychiatric disorder
 - Accounted for differences in overall survival according to psychiatric disorder and presence of TD symptoms

METHODS

MODEL OVERVIEW

- A discretely integrated condition event (DICE) model[6] was used to evaluate clinical and economic outcomes associated with VBZ and DTBZ treatment of TD
- The model conceptualizes "conditions" that reflect aspects of the model or patient attributes that persist, and "events" that reflect points in time when conditions may change (**Figure 1**)

Figure 1. Discretely Integrated Condition Event Model

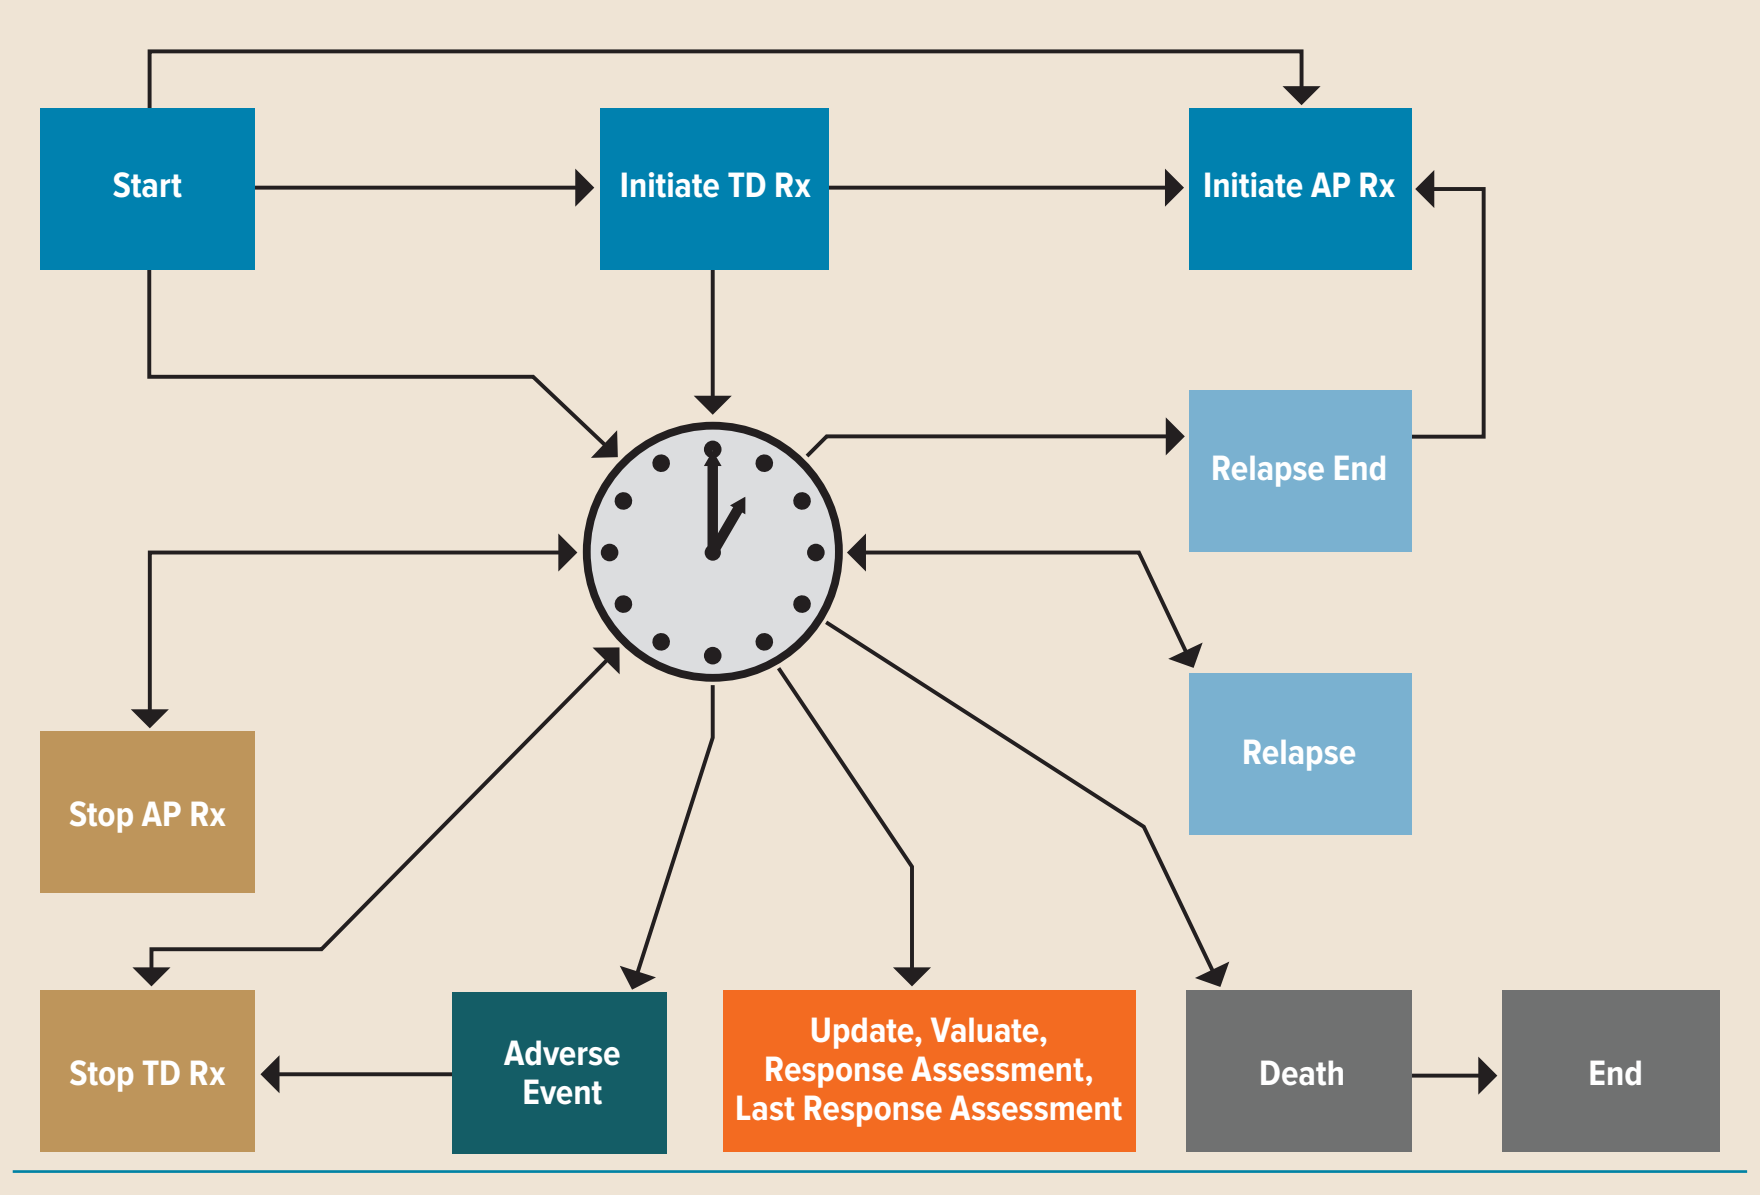
Start
Initiate TD Rx
Initiate AP Rx
Relapse End
Relapse
Stop AP Rx
Stop TD Rx
Adverse Event
Update, Valuate, Response Assessment, Last Response Assessment
Death
End

Times to events are initially determined at the start of the model. These times are periodically revised during Update, Valuate, Response Assessment, and Last Response Assessment events, which are part of the scheduling process represented by the clock, and when other events occur, as indicated by arrowheads.
AP, antipsychotic; Rx, prescription; TD, tardive dyskinesia.

PATIENT POPULATION AND PERSPECTIVE

- A synthetic population of 1,000 patients with demographic and clinical characteristics derived from population statistics in the KINECT 3 trial of VBZ[7] was created and used in the model
- Patients could have only one underlying psychiatric condition: schizophrenia, bipolar disorder, or major depressive disorder (MDD)
- The model was analyzed from a U.S. third-party payer perspective over a lifetime horizon
 - Only direct costs (drug acquisition costs, disease management costs, and relapse treatment costs) were included in the analysis

MODEL INPUTS

- Clinical inputs were taken primarily from published literature (**Table 1**)

Table 1. Clinical Inputs

Average Daily TD Medication Doses[a]

Drug/Dose	Proportion of Patients	Source
VBZ 40 mg	0.200	Factor et al. *J Clin Psychiatry*; 2017 Marder et al. *Neruopsychopharmacol*; 2017 (Suppl) Anderson et al. *Lancet Psychiatry*; 2017 Fernandez et al. *Neurology*; 2017 Hauser et al. *Neurology*; 2018 (15 Suppl)
VBZ 80 mg	0.800	
DTBZ 24 mg	0.270	
DTBZ 36 mg	0.270	
DTBZ 48 mg	0.460	

Probability of Response Measured by Global Response (CGIC score ≤2, PGIC score ≤2)

Week	VBZ 40 mg	VBZ 80 mg	DTBZ	Source
Week 8	0.425, 0.457	0.473, 0.441	0.410, 0.390	Factor et al. *J Clin Psychiatry*; 2017 Marder et al. *Neruopsychopharmacol*; 2017 (Suppl) Anderson et al. *Lancet Psychiatry*; 2017 Fernandez et al. *Neurology*; 2017 Hauser et al. *Neurology*; 2018 (15 Suppl)
Week 16	0.559, 0.577	0.595, 0.622	0.440, 0.440	
Week 24	0.648, 0.758	0.821, 0.772	0.620, 0.590	
Week 48	0.667, 0.741	0.869, 0.832	0.680, 0.660	

Parameters for Antipsychotic Treatment Discontinuation

Parameter, Patient Type	Schizophrenia	Bipolar Disorder	MDD	Source
Weibull parameters (γ, λ), patients with TD	0.783, 0.952	0.914, 1.268	0.914, 1.268	Greene et al. *J Med Econ*; 2018 Prater et al. *J Manag Care Spec Pharm*; 2018 Institute for Clinical and Economic Review; 2017
Hazard ratio, TD responder	0.593	0.593	0.593	

Annual Risk for Psychiatric Relapse by Antipsychotic Medication Status

Medication Status	Schizophrenia	Bipolar Disorder	MDD	Source
On medication	0.315	0.528	0.528	Leucht et al. *Lancet*; 2012 Derry et al. *BMC Psychiatry*; 2007 Di Capite et al. *Early Interv Psychiatry*; 2018
Off medication	1.022	1.050	1.050	

Psychiatric Relapse Sequelae

Parameter	Schizophrenia	Bipolar Disorder	MDD	Source
Proportion hospitalized	0.564	0.564	0.564	Panish et al. *J Pharm Health Serv Res*; 2013 Park et al. *Value Health*; 2014 Rajagopalan et al. *J Med Econ*; 2013 Ascher-Svanum et al. *BMC Psychiatry*; 2010
Duration of hospitalization, days	10.7	6.9	6.5	
Duration of outpatient treatment, days	9.0	9.0	9.0	

Hazard Ratio for Mortality by Psychiatric Condition

Gender	Schizophrenia	Bipolar Disorder	MDD	Source
Male	2.210	1.750	0.880	Crump et al. *JAMA Psychiatry*; 2013 Chiu et al. *J Affect Disord*; 2018
Female	2.660	2.090	2.140	

[a]Average cohort dose: 72.0 mg/day for VBZ, 38.3 mg/day for DTBZ.
CGIC, Clinical Global Impression of Change; DTBZ, deutetrabenazine; MDD, major depressive disorder; PGIC, Patient Global Impression of Change; TD, tardive dyskinesia; VBZ, valbenazine.

- The mean utilities for baseline psychiatric conditions of schizophrenia, bipolar disorder, and MDD were 0.83, 0.80, and 0.80, respectively
- A utility decrement of 0.143 was applied to reflect moderate to severe TD
- Utility decrements of 0.081, 0.118, and 0.118 were applied to reflect relapses in schizophrenia, bipolar disorder, and MDD, respectively
- Mortality was modeled using general population estimates adjusted to reflect condition-specific hazard ratios of the underlying psychiatric disorders
- The hazard ratio for mortality used for patients with TD was 1.900
- Cost inputs (in 2017 U.S. dollars) are shown in **Table 2**
- Costs, quality-adjusted life years (QALYs), and life years were discounted at an annual rate of 3%

KEY ASSUMPTIONS

- All patients began with moderate to severe TD, having attempted optimization or titration of their antipsychotic medications
- During the first 24 weeks of treatment with VBZ, DTBZ, or no treatment, patients were assessed every 8 weeks for response, defined as a Clinical Global Impression of Change (CGIC) score ≤2 or a Patient Global Impression of Change (PGIC) score ≤2
 - Patients who responded at a given assessment suffered no disutility due to TD until at least the next assessment
- A final response assessment was conducted at Week 48, at which time responders continued TD treatment until death, and non-responders discontinued TD treatment
- Patients who discontinued one TD treatment did not attempt another
- All patients with a psychiatric diagnosis were at risk of discontinuing their antipsychotic medications, putting them at increased risk of relapse
 - Patients who respond to treatment were assumed to have reduced symptoms of TD, which improves adherence to antipsychotic treatment and lowers the risk of relapse
 - Patients receiving antipsychotic medication were assumed to discontinue upon relapse
 - Antipsychotic medication was restarted at the end of the relapse
- The primary health outcomes assessed were QALYs, life years, response to TD treatment at 1 year, and number of relapses

Table 2. Tardive Dyskinesia Treatment Cost Inputs

Daily Medication Costs

Drug	Daily Dose (mg)[a]	Daily Cost (2017 US $)	Source
VBZ	40	139.92	For VBZ and DTBZ, based on 27% discount to Wholesale Acquisition Cost For Antipsychotic, Gilmer et al. *Am J Psychiatry*; 2004
VBZ	80	151.48	
DTBZ	12	83.94	
DTBZ	18	94.42	
DTBZ	24	125.90	
DTBZ	30	178.35	
DTBZ	36	188.84	
DTBZ	42	220.31	
DTBZ	48	251.79	
Antipsychotic	—	17.35	

Annual Disease Management Costs

Disorder	Cost with TD (2017 US $)	Cost without TD (2017 US $)	Source
Schizophrenia	7,909.94	5,361.92	Gilmer et al. *Am J Psychiatry*; 2004 Guo et al. *Value Health*; 2008
Bipolar disorder	3,983.06	1,435.04	
MDD	3,983.06	1,435.04	
No psychiatric disorder	2,548.02	—	

Psychiatric Disorder Relapse Costs

Disorder	Hospital Costs (2017 US $)	Outpatient Costs (2017 US $)	Source
Schizophrenia	853.72	84.13	Park et al. *Value Health*; 2014
Bipolar disorder	839.05	84.13	
MDD	546.08	84.13	

[a]Average cohort dose: 72.0 mg/day for valbenazine, 38.3 mg/day for deutetrabenazine.
DTBZ, deutetrabenazine; MDD, major depressive disorder; TD, tardive dyskinesia; VBZ, valbenazine.

SENSITIVITY ANALYSES

- Deterministic sensitivity analyses assessed the impact of varying drug acquisition costs, probabilities of response, risk of relapse, and hazard ratio for TD mortality by ±20%
- Probabilistic sensitivity analyses that drew random values from the distributions of all the model parameters assessed the likelihood of cost-effectiveness of each intervention relative to a range of willingness-to-pay thresholds

RESULTS

BASE CASE ANALYSIS

- Lifetime health and total cost outcomes for patients with moderate to severe TD who received treatment with a VMAT2 inhibitor or no treatment are shown in **Table 3** and **Table 4**
- Patients treated with VBZ or DTBZ showed reduced TD symptoms, increased survival and QALYs, and decreased disease management costs compared with no treatment
- Whether measured by clinicians or patients, assessments of global improvement resulted in remarkably consistent health and cost outcomes for each VMAT2 inhibitor
- Regardless of the response criterion, VBZ dominated DTBZ with higher probability of response at 1 year, more QALYs, and lower lifetime costs

Table 3. Lifetime Health and Cost Outcomes of Tardive Dyskinesia Treatment

Outcome	VBZ	DTBZ	No Treatment	VBZ vs. No Treatment	VBZ vs. DTBZ
Response Criterion: CGIC score ≤2					
QALYs (discounted)	10.232	9.704	7.163	3.069	0.528
LYs (discounted)	12.843	12.453	10.588	2.255	0.390
Responders at year 1, %	79.1	65.5	0	—	—
Relapses, n	10.8	10.6	9.3	—	—
Total discounted costs (2017 US $)	748,053	828,298	139,578	608,475	(80,244)
ICER/QALY	—	—	—	198,243	Dominant
Response Criterion: PGIC score ≤2					
QALYs (discounted)	10.185	9.661	7.163	3.022	0.524
LYs (discounted)	12.811	12.422	10.588	2.222	0.389
Responders at year 1, %	78.0	64.4	0	—	—
Relapses, n	10.8	10.6	9.3	—	—
Total discounted costs (2017 US $)	737,389	818,045	139,578	597,811	(80,656)
ICER/QALY	—	—	—	197,825	Dominant

CGIC, Clinical Global Impression of Change; DTBZ, deutetrabenazine; ICER, incremental cost-effectiveness ratio; LY, life year; PGIC, Patient Global Impression of Change; QALY, quality-adjusted life year; VBZ, valbenazine.

Table 4. Lifetime Discounted Costs of Tardive Dyskinesia Treatment

Cost Component	VBZ (2017 US $)	DTBZ (2017 US $)	No Treatment (2017 US $)
Response Criterion: CGIC score ≤2			
TD medication	601,360	682,969	—
AP medication	51,008	48,203	34,561
Disease management	59,237	61,277	71,349
Disease relapse	36,448	35,849	33,668
Total	748,053	828,298	139,578
Response Criterion: PGIC score ≤2			
TD medication	590,794	672,845	—
AP medication	50,695	47,964	34,561
Disease management	59,486	61,453	71,349
Disease relapse	36,414	35,784	33,668
Total	737,389	818,045	139,578

AP, antipsychotic; CGIC, Clinical Global Impression of Change; DTBZ, deutetrabenazine; PGIC, Patient Global Impression of Change; TD, tardive dyskinesia; VBZ, valbenazine.

SENSITIVITY ANALYSES

- Tornado plots show the effects of varying model inputs by ±20% for VBZ compared with no treatment (**Figure 2**) and compared with DTBZ (**Figure 3**), using the PGIC response criterion
 - The results were most sensitive to the acquisition cost of VBZ 80 mg
 - VBZ dominated DTBZ when varying most model inputs by ±20%
- The cost-effectiveness acceptability curves derived from the probabilistic sensitivity analyses using the PGIC response criterion are shown in **Figure 4**
 - At a cost threshold of approximately $200,000 per QALY, VBZ is more likely than not to be cost effective compared with no treatment
 - DBTZ is not a cost-effective option at any willingness-to-pay threshold ≤$300,000 per QALY
- Similar results for the sensitivity analyses were obtained using the CGIC response criterion

Figure 2. Tornado Plots for Deterministic Sensitivity Analysis of Valbenazine Compared with No Treatment

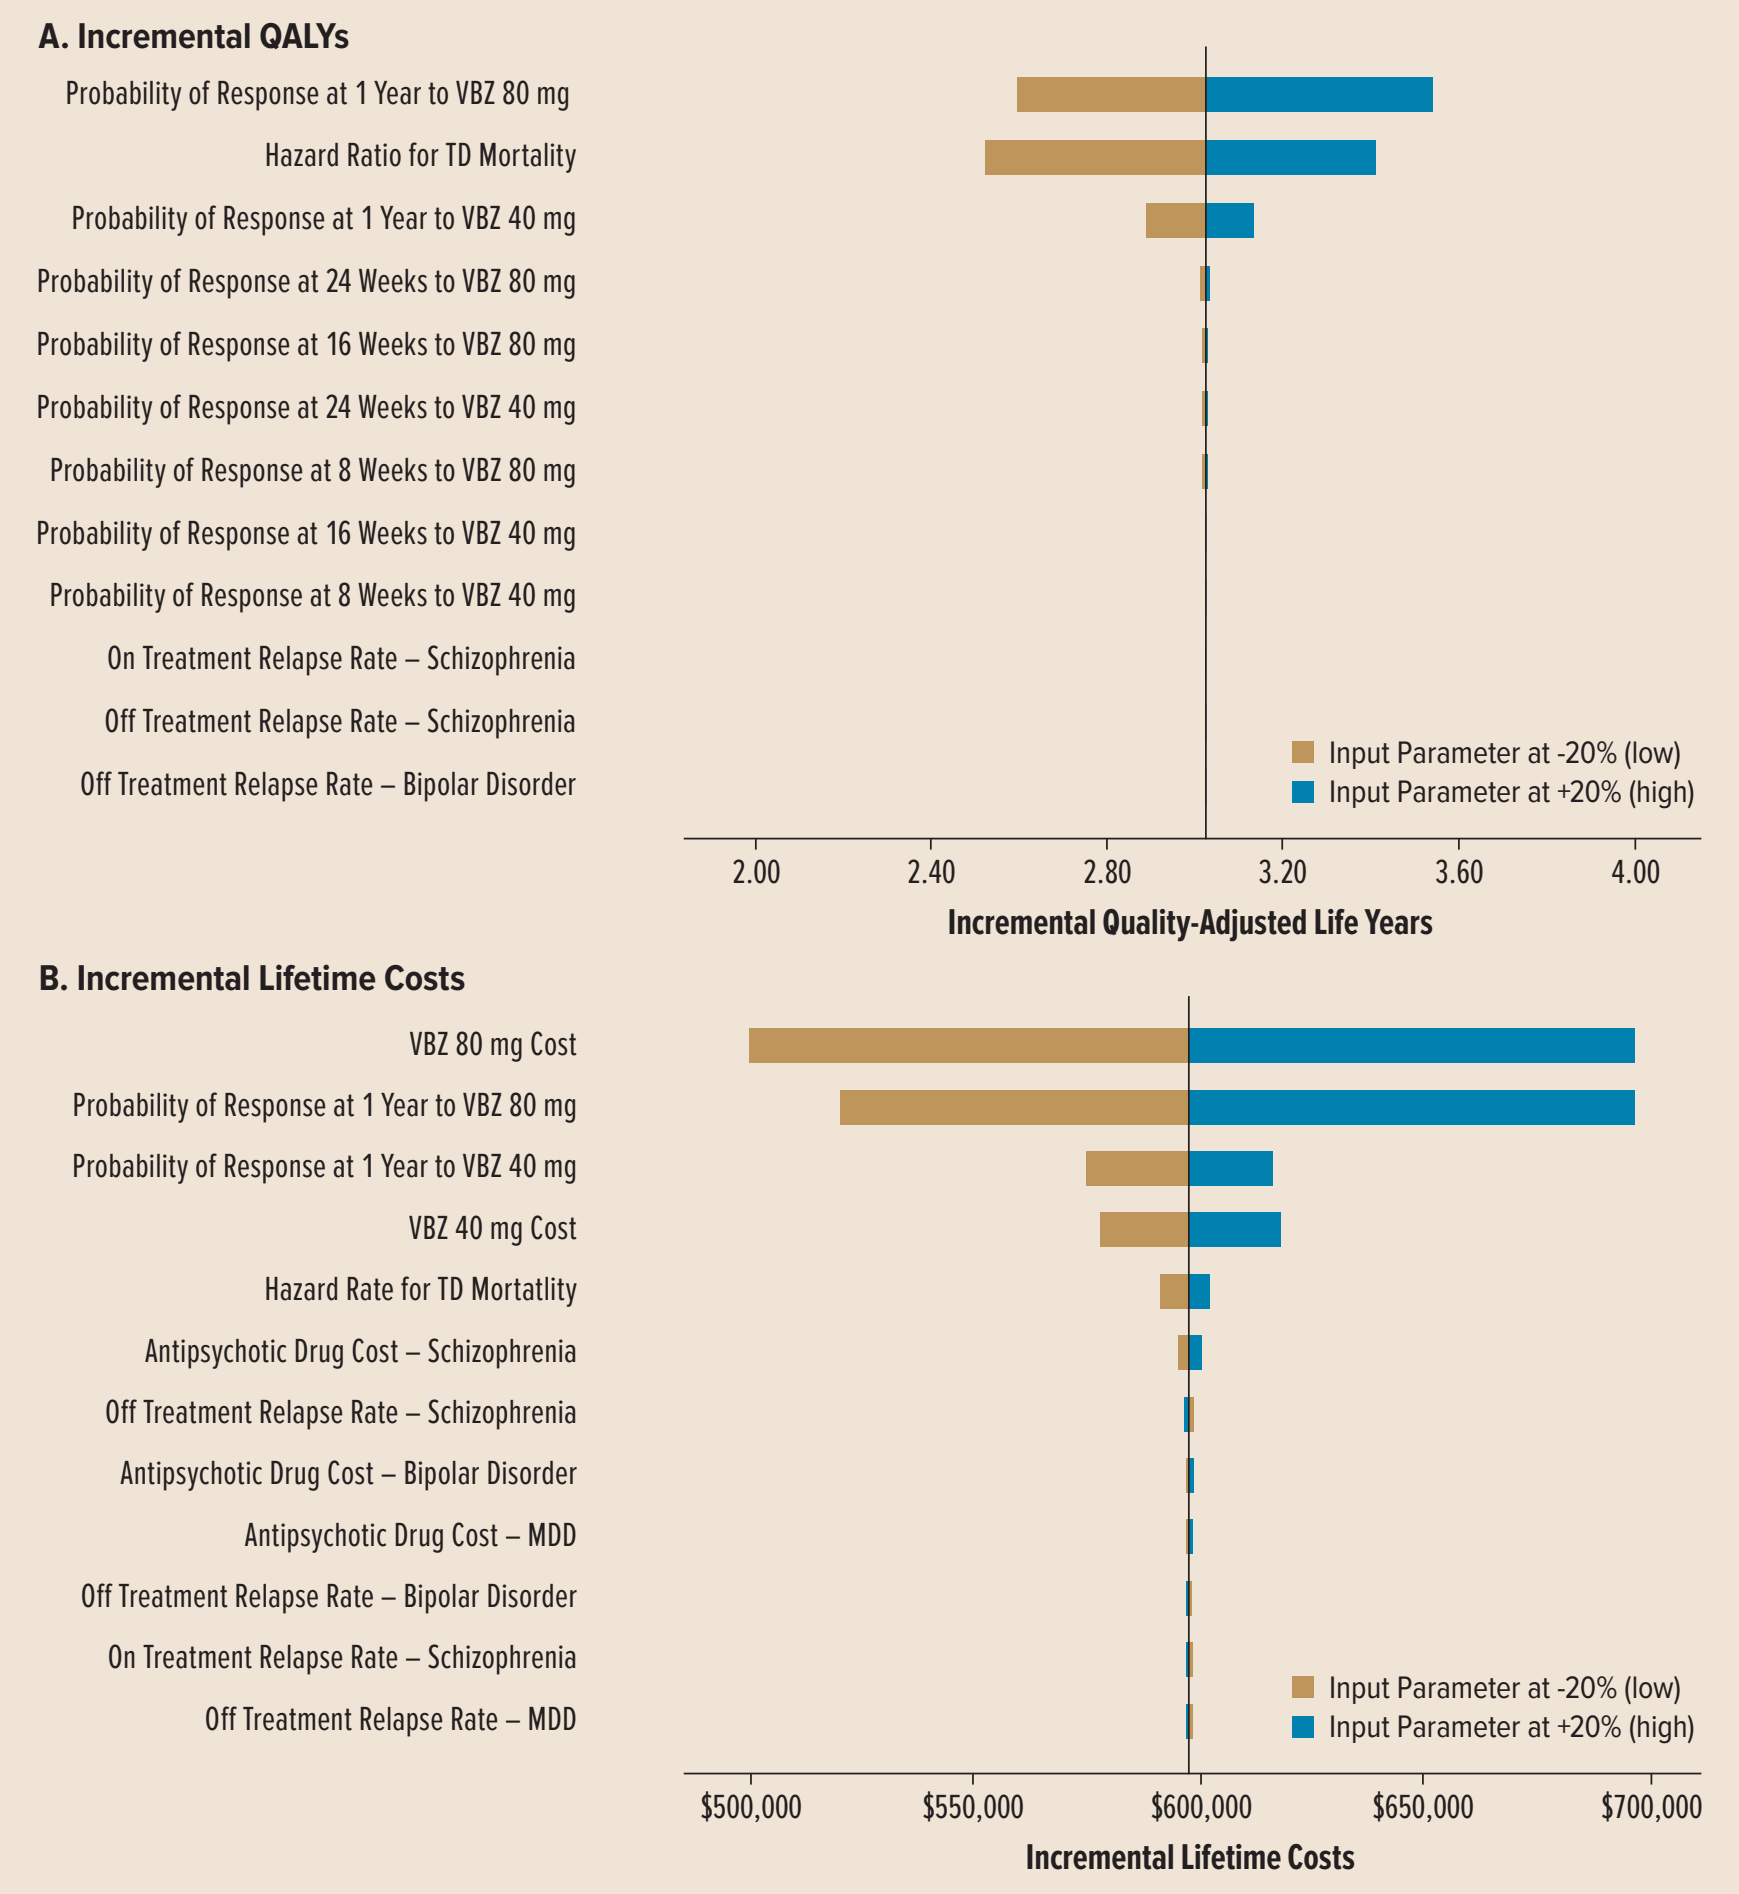
A. Incremental QALYs
Probability of Response at 1 Year to VBZ 80 mg
Hazard Ratio for TD Mortality
Probability of Response at 1 Year to VBZ 40 mg
Probability of Response at 24 Weeks to VBZ 80 mg
Probability of Response at 16 Weeks to VBZ 80 mg
Probability of Response at 24 Weeks to VBZ 40 mg
Probability of Response at 8 Weeks to VBZ 80 mg
Probability of Response at 16 Weeks to VBZ 40 mg
Probability of Response at 8 Weeks to VBZ 40 mg
On Treatment Relapse Rate – Schizophrenia
Off Treatment Relapse Rate – Schizophrenia
Off Treatment Relapse Rate – Bipolar Disorder
Input Parameter at -20% (low)
Input Parameter at +20% (high)
2.00
2.40
2.80
3.20
3.60
4.00
Incremental Quality-Adjusted Life Years
B. Incremental Lifetime Costs
VBZ 80 mg Cost
Probability of Response at 1 Year to VBZ 80 mg
Probability of Response at 1 Year to VBZ 40 mg
VBZ 40 mg Cost
Hazard Rate for TD Mortatlity
Antipsychotic Drug Cost – Schizophrenia
Off Treatment Relapse Rate – Schizophrenia
Antipsychotic Drug Cost – Bipolar Disorder
Antipsychotic Drug Cost – MDD
Off Treatment Relapse Rate – Bipolar Disorder
On Treatment Relapse Rate – Schizophrenia
Off Treatment Relapse Rate – MDD
Input Parameter at -20% (low)
Input Parameter at +20% (high)
$500,000
$550,000
$600,000
$650,000
$700,000
Incremental Lifetime Costs

Analysis based on PGIC score ≤2 response criterion.
MDD, major depressive disorder; PGIC, Patient Global Impression of Change; QALY, quality-adjusted life year; TD, tardive dyskinesia; VBZ, valbenazine.

Figure 3. Tornado Plots for Deterministic Sensitivity Analysis of Valbenazine Compared with Deutetrabenazine

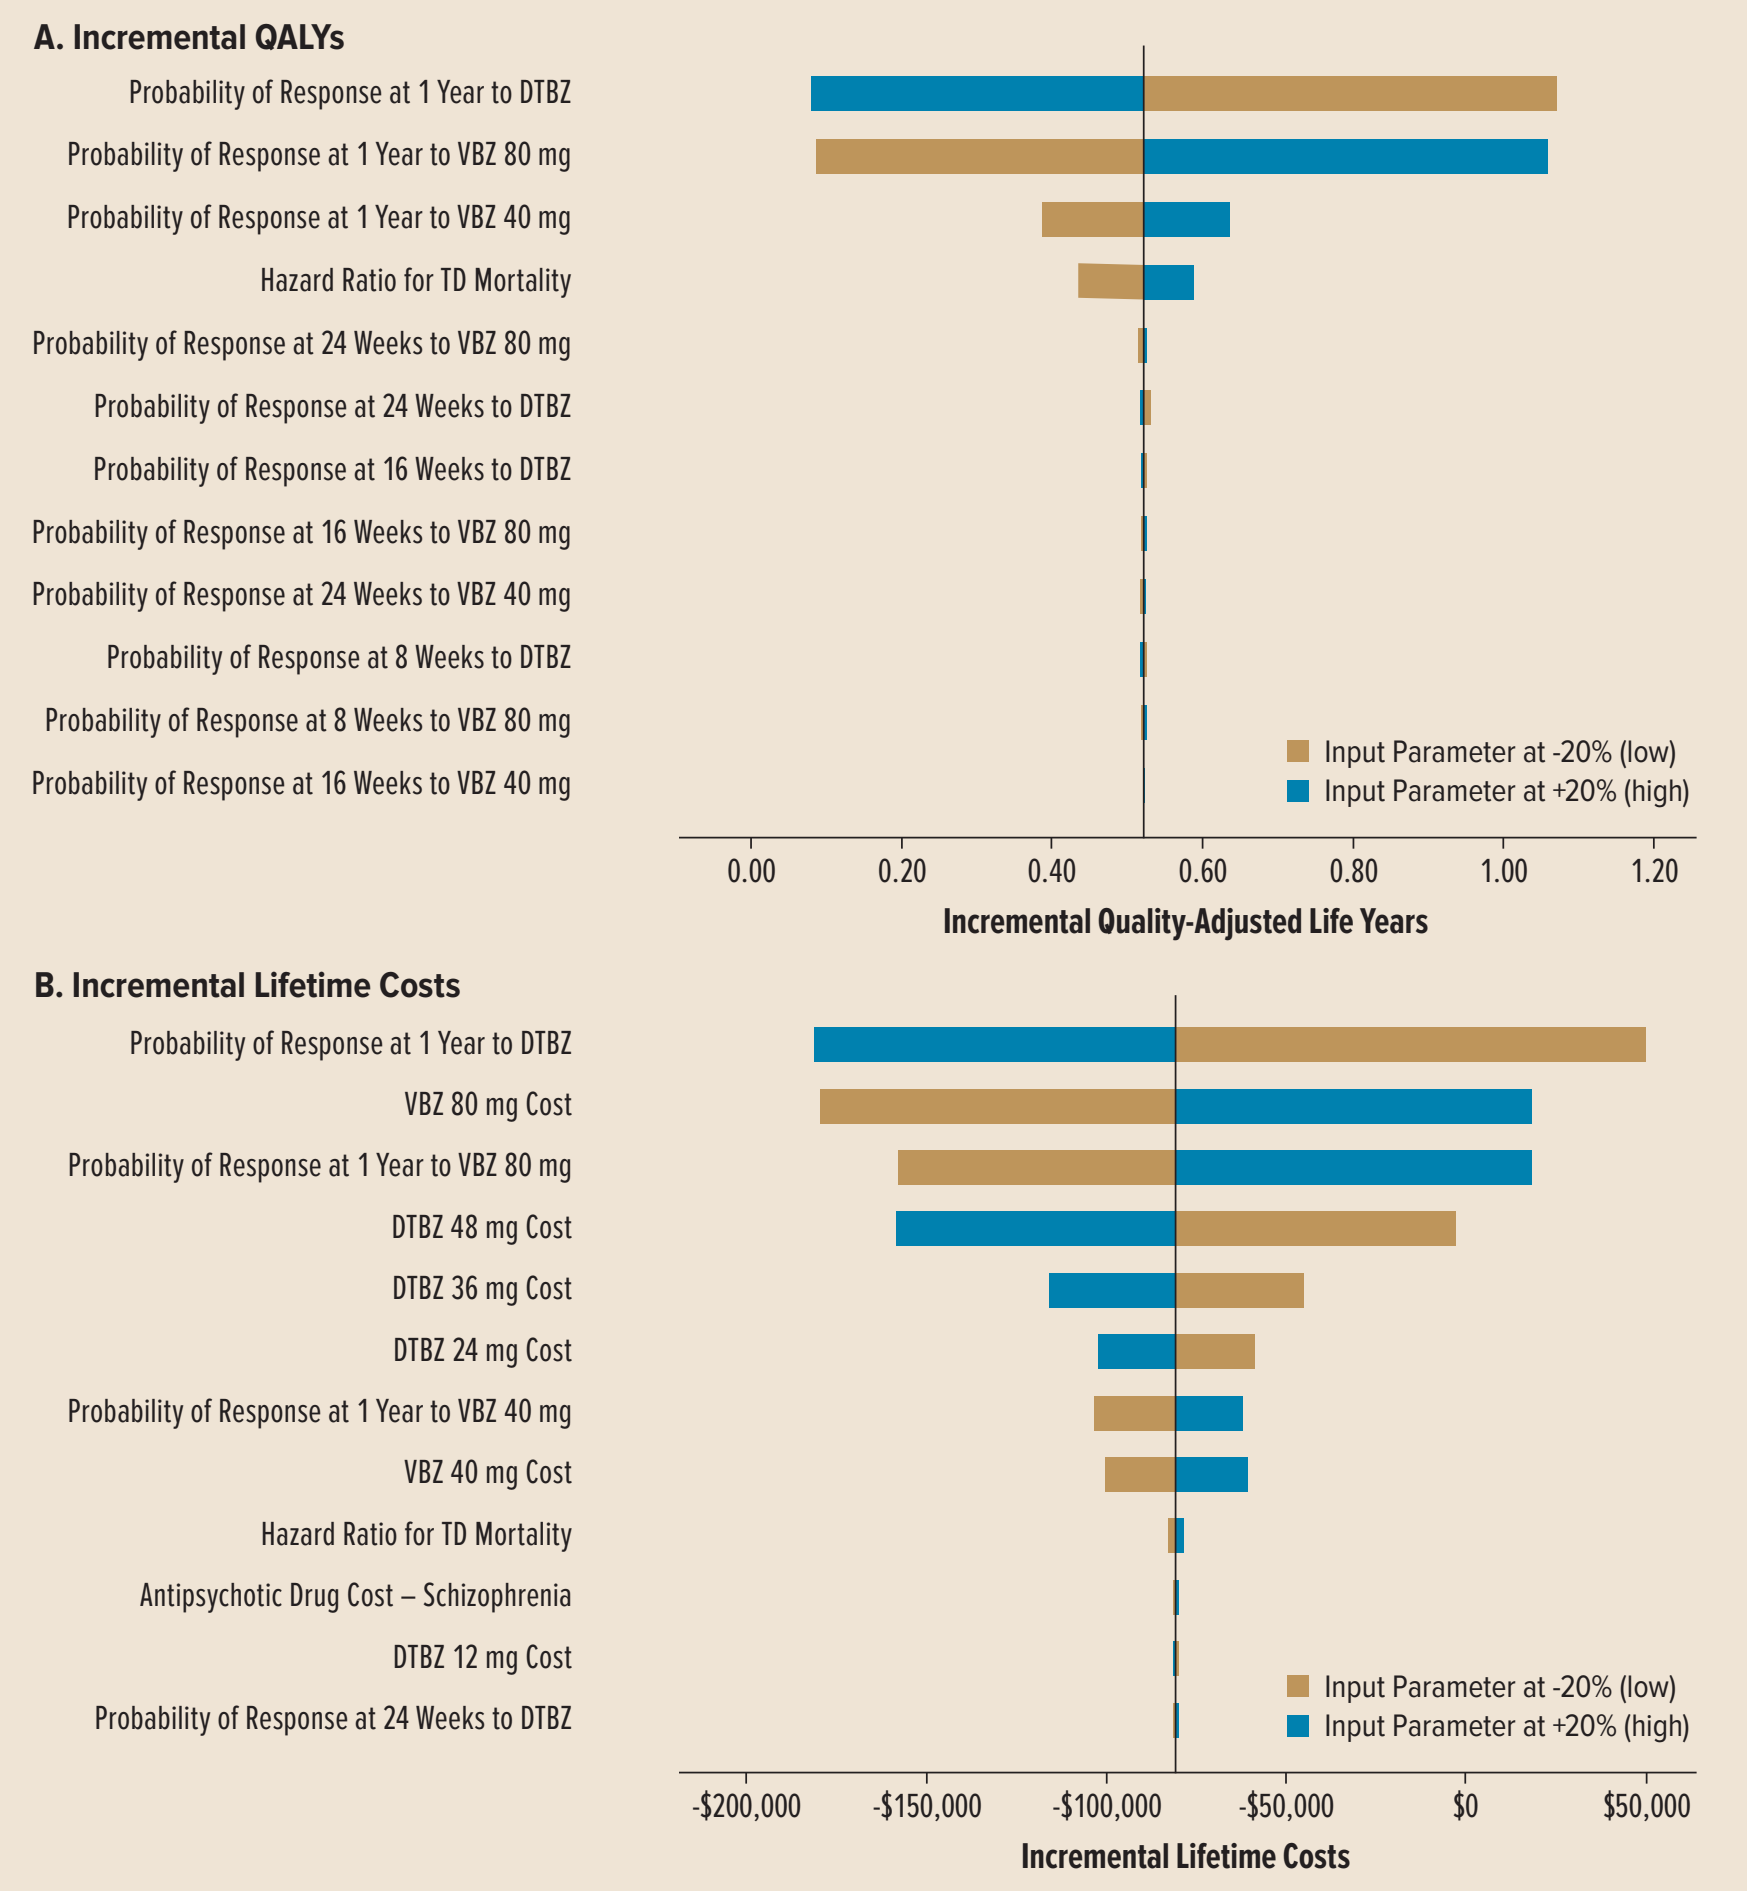
A. Incremental QALYs
Probability of Response at 1 Year to DTBZ
Probability of Response at 1 Year to VBZ 80 mg
Probability of Response at 1 Year to VBZ 40 mg
Hazard Ratio for TD Mortality
Probability of Response at 24 Weeks to VBZ 80 mg
Probability of Response at 24 Weeks to DTBZ
Probability of Response at 16 Weeks to DTBZ
Probability of Response at 16 Weeks to VBZ 80 mg
Probability of Response at 24 Weeks to VBZ 40 mg
Probability of Response at 8 Weeks to DTBZ
Probability of Response at 8 Weeks to VBZ 80 mg
Probability of Response at 16 Weeks to VBZ 40 mg
Input Parameter at -20% (low)
Input Parameter at +20% (high)
0.00
0.20
0.40
0.60
0.80
1.00
1.20
Incremental Quality-Adjusted Life Years
B. Incremental Lifetime Costs
Probability of Response at 1 Year to DTBZ
VBZ 80 mg Cost
Probability of Response at 1 Year to VBZ 80 mg
DTBZ 48 mg Cost
DTBZ 36 mg Cost
DTBZ 24 mg Cost
Probability of Response at 1 Year to VBZ 40 mg
VBZ 40 mg Cost
Hazard Ratio for TD Mortality
Antipsychotic Drug Cost – Schizophrenia
DTBZ 12 mg Cost
Probability of Response at 24 Weeks to DTBZ
Input Parameter at -20% (low)
Input Parameter at +20% (high)
-$200,000
-$150,000
-$100,000
-$50,000
$0
$50,000
Incremental Lifetime Costs

Analysis based on PGIC score ≤2 response criterion.
DTBZ, deutetrabenazine; MDD, major depressive disorder; PGIC, Patient Global Impression of Change; QALY, quality-adjusted life year; TD, tardive dyskinesia; VBZ, valbenazine.

Figure 4. Cost-Effectiveness Acceptability Curves

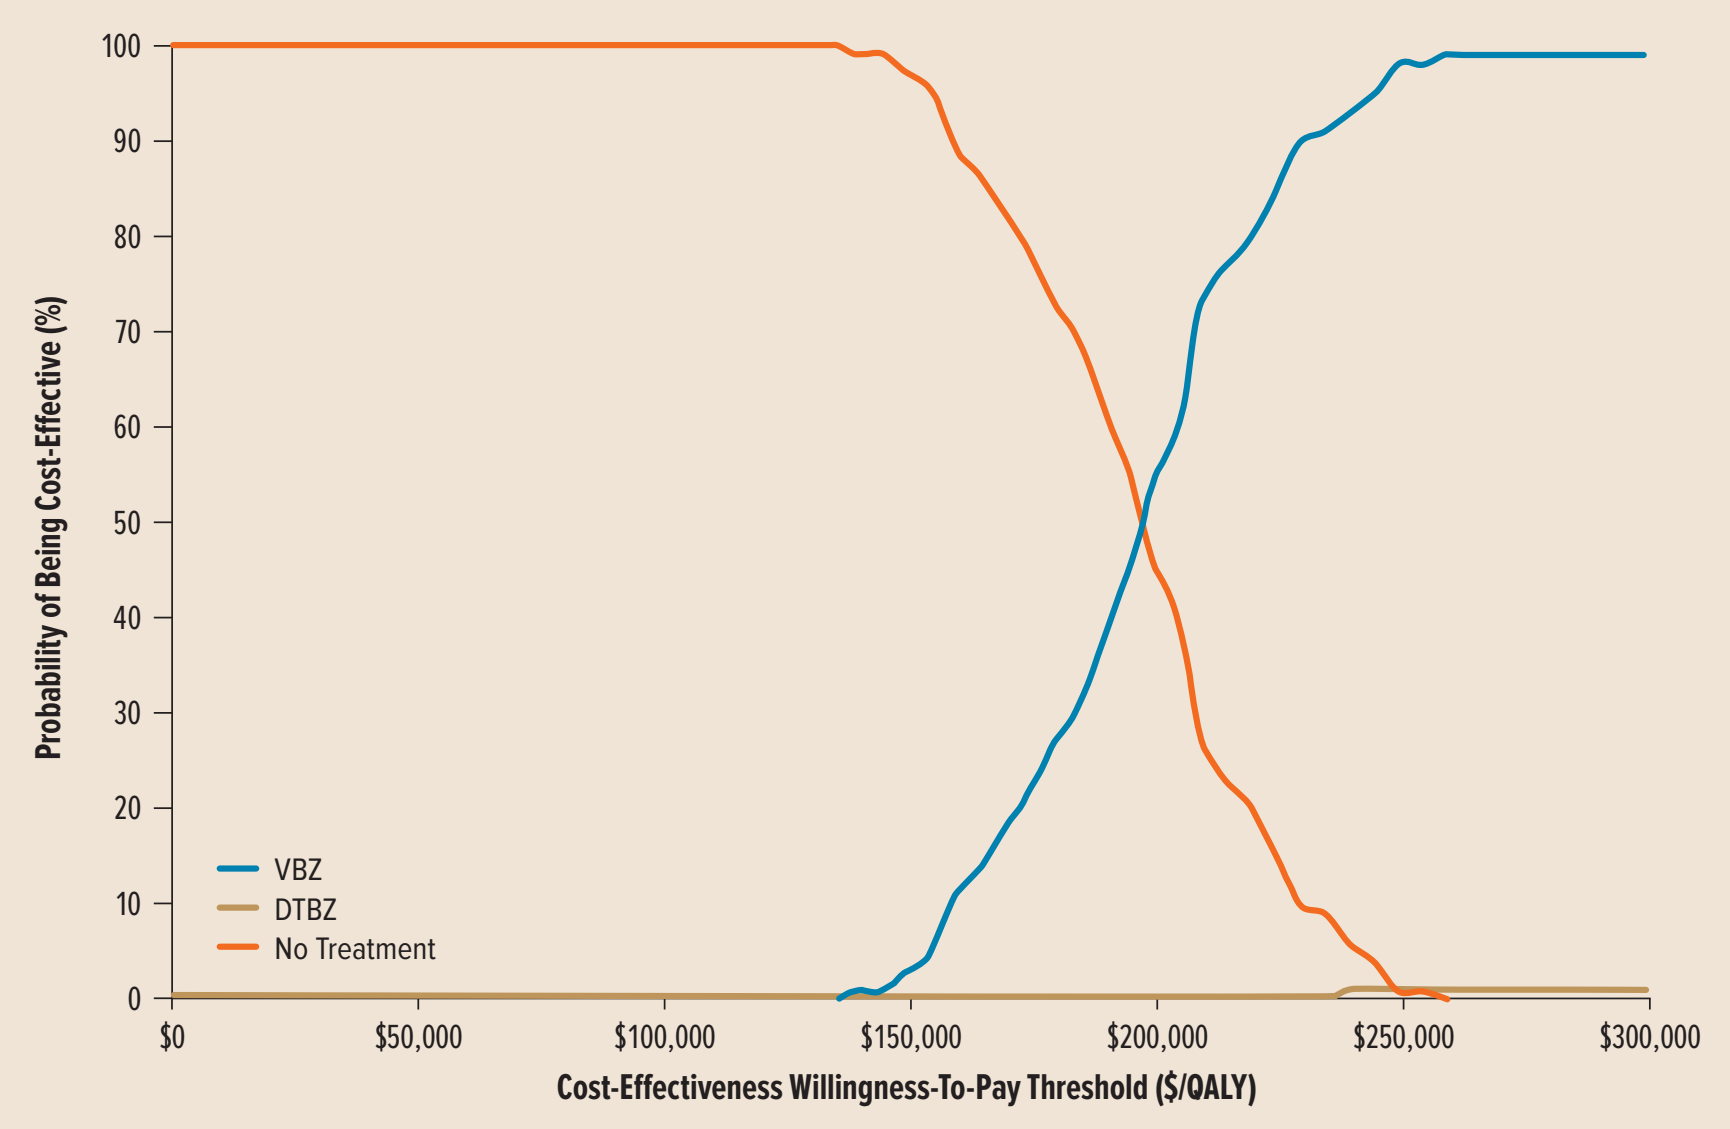
100
90
80
70
60
50
40
30
20
10
0
Probability of Being Cost-Effective (%)
VBZ
DTBZ
No Treatment
$0
$50,000
$100,000
$150,000
$200,000
$250,000
$300,000
Cost-Effectiveness Willingness-To-Pay Threshold ($/QALY)

Analysis based on PGIC score ≤2 response criterion.
DTBZ, deutetrabenazine; PGIC, Patient Global Impression of Change; QALY, quality-adjusted life year; VBZ, valbenazine.

CONCLUSIONS

- In this model, treatment with VBZ and DTBZ was associated with longer life expectancy and better quality of life compared with no treatment in patients with moderate to severe TD
- The patient perspective of global improvement may be most relevant when determining the value of treatment for TD
- Patients using VBZ accrued more lifetime QALYs and incurred lower lifetime costs than patients using DTBZ
- Results of this study will assist payers in making more fully informed decisions regarding treatment of TD

REFERENCES

1. Waln O, Jankovic J. *Tremor Other Hyperkinet Mov (N.Y.)*. 2013;3.
2. Cloud LJ, et al. *Neurotherapeutics*. 2014;11:166-76.
3. Zutshi D, et al. *Tremor Other Hyperkinet Mov (N.Y.)*. 2014;4:266.
4. Browne S, et al. *Acta Psychiatr Scand*. 1996;94:118-24.
5. Rosenheck RA. *Br J Psychiatry*. 2007;191:238-45.
6. Caro JJ. *Pharmacoeconomics*. 2016;34:665-72.
7. Hauser RA, et al. *Am J Psychiatry*. 2017;174:476-84.

Disclosures: This study was supported by Neurocrine Biosciences, Inc., San Diego, CA. Writing assistance and editorial support was provided by Prescott Medical Communications Group, Inc., Chicago, IL.

PRESENTED AT THE 24TH ANNUAL INTERNATIONAL SOCIETY FOR PHARMACOECONOMICS OUTCOMES RESEARCH MEETING
MAY 18-22, 2019; NEW ORLEANS, LA